Cost-Effectiveness and Budget Impact Analyses of Implementing the First Pillar of Patient Blood Management with Intravenous (IV) Ferric Carboxymaltose (FCM) on the Turkish Healthcare System

Ramirez de Arellano Serna A[1], Tatar M[2], Alkis N[2], Guclu CY[2], Bermede O[2], Erdemli B[2], Gunaydin S[3]

[1]CSL Vifor, Glattbrugg, ZH, Switzerland, [2]Polar Health Economics and Policy, Ankara, Turkey, Ankara, Turkey, [3]Ankara City Hospital, University of Health Sciences, Ankara, Turkey

EE562

INTRODUCTION

- Patient blood management (PBM) is a patient-centered, evidence-based multidisciplinary approach that aims to optimize hemoglobin concentration, maintain hemostasis and minimize blood loss in patients undergoing surgery.[1-3]
- The available evidence indicates that anemia, bleeding and exposure to allogeneic products are risk factors for morbidity and mortality in elective surgery.[4-8]
- These risk factors account for an economic burden on healthcare systems through prolonged length of stay in hospital (LOS), re-hospitalizations and increased risk of adverse events and complications.
- The Ministry of Health (MoH) of Turkey embarked on a project titled '*Technical Assistance for Improving Blood Transfusion Management in Turkey*' in March 2019 with assistance from the European Union.[9]

OBJECTIVES

- The aims of this study is: (1) to explore the **cost-effectiveness** of comprehensive anemia management, first pillar of PBM, in non-cardiac and cardiac surgery from the Turkish Social Security Institution (SSI) perspective; (2) to explore the **budget impact** of PBM for coronary artery bypass grafting and hip & knee arthroplasty to the SSI.

METHODS

- A decision tree model with probabilities of adverse events was developed to assess the cost-effectiveness of PBM versus no PBM in non-cardiac (hip & knee arthroplasties – H&K) and cardiac surgeries (coronary artery bypass grafting –CABG) with a simulated cohort of 10,000 patients in Turkey (**Figure 1**).
- The endpoints of the study were postoperative adverse events avoided for a hospitalization period of up to 30 days (sepsis with or without pneumonia, acute renal failure, acute myocardial infarction and acute stroke). The implementation of the first pillar of PBM (i.e. preoperative anemia measure) was compared with placebo.
- Preoperative anemia treatment was made with intravenous (IV) ferric carboxymaltose (FCM) (two 500 mg per 10 mL vials before surgery).
- Data on endpoints and probabilities for non-cardiac and cardiac surgeries were obtained from the results of the Kleinerüschkamp et al. study,[8] and given as incremental cost per avoided postoperative complication.
- The budget impact analysis was based on the costs of treating postoperative adverse events and the cost of receiving PBM. **Figure 2** shows the epidemiological and costing data used in the analysis.
- In 2017, there were 12,237 CABG surgeries and 77,780 hip and knee arthroplasties in Turkish MoH hospitals. According to Ünal et al (2020)[10], 33% of these would have preoperative anemia (4038 patients undergoing CABG and 25,667 patients with H&K arthroplasty). Based on Drabinski et al (2021)[11], 50% of the patients with preoperative anemia would have iron deficiency anemia (IDA) (2019 patients undergoing CABG and 12,834 patients with H&K arthroplasty). Finally, on the basis of expert opinion, 50% of patients with IDA would receive 1000 mg of IV FCM treatment (1010 patients undergoing CABG and 6417 patients with H&K arthroplasty).
- Sensitivity analysis for both CEA and BIM were conducted to check the robustness of results.

RESULTS

- PBM was found to dominate the control arm in both non-cardiac and cardiac surgeries and provided better outcomes with lower costs (**Table 2**). With the implementation of PBM, the incremental cost was –12,122 TRY for non-cardiac surgery and –11,449 TRY for cardiac surgery.
- The overall net cost savings related to avoided post-surgical adverse events following hip and knee arthroplasty in Turkey in 2017, were 70,729,809 TRY (€3,844,011) (**Table 3**). For CABG surgeries were 35,979,085 TRY (€1,955,385).

CONCLUSIONS

- In our study, the implementation of PBM was associated with a decreased rate of adverse events in both cardiac and non-cardiac surgical patients. PBM should be advocated as a cost-effective and cost-saving option in major surgeries in Turkey. The SSI can play a leading role by promoting, regulating and implementing policy for the inclusion of PBM in hospital-based process improvement initiatives with the goal of improving patient safety and clinical outcomes.

Figure 1. Decision tree model comparing PBM versus no PBM implementation

Decision Tree Cost-effectiveness Analysis

First decision node:
PBM versus control (no PBM)
Probability nodes:
1. probability for adverse events
2. probabilities for type of event
Terminal nodes:
Avoided adverse events & associated costs

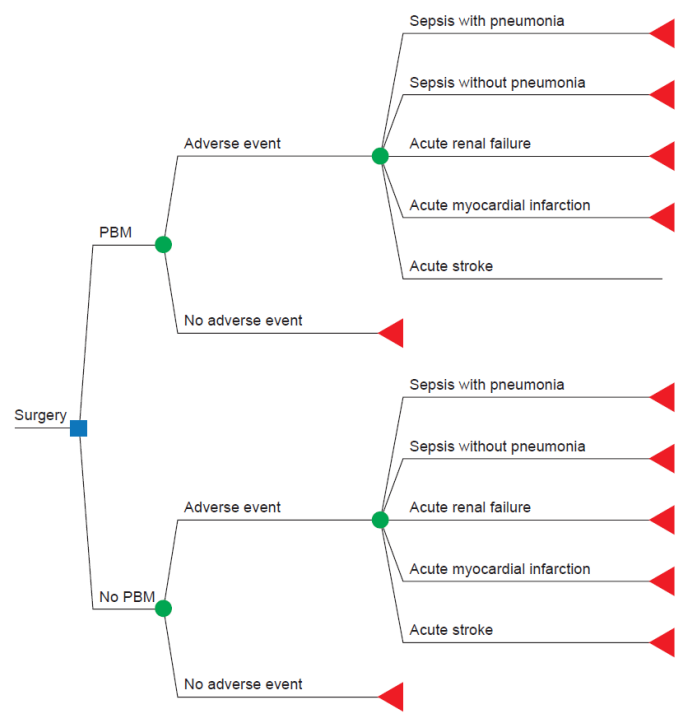
Surgery
PBM
No PBM
Adverse event
No adverse event
Sepsis with pneumonia
Sepsis without pneumonia
Acute renal failure
Acute myocardial infarction
Acute stroke

Table 1. Postoperative adverse events and probabilities for non-cardiac & cardiac surgeries

	Control arm			PBM arm			Total avoided adverse events
	n	%	Probability	n	%	Probability	n
Sepsis with pneumonia	1108	25.14	0.2514	156	3.54	0.0354	952
Sepsis without pneumonia	824	18.69	0.1869	416	9.44	0.0944	408
Acute renal failure	402	9.12	0.0912	198	4.49	0.0449	204
Acute MI	596	13.52	0.1352	450	10.21	0.1021	146
Acute stroke	158	3.58	0.0358	100	2.27	0.0227	58
Total adverse events	**3088**			**1320**			**1768**

	Control arm			PBM arm			Total avoided adverse events
	n	%	Probability	n	%	Probability	n
Sepsis with pneumonia	648	25.89	0.2590	265	10.59	0.1059	383
Sepsis without pneumonia	482	19.26	0.1926	197	7.87	0.0787	285
Acute renal failure	250	9.99	0.0999	94	3.76	0.0376	156
Acute MI	303	12.11	0.1211	7	0.28	0.0028	296
Acute stroke	190	7.59	0.0759	66	2.64	0.0264	124
Total adverse events	**1873**			**629**			**1244**

Figure 2. Budget Impact Analysis: epidemiological & costing data in Turkey

Budget Impact Analysis

- 33% of patients with preoperative anemia[1]
- 50% of patients with preoperative anemia have IDA[3]
- 50% of patients with IDA treated with FCM
- 1000 mg IV FCM (two vials) per patient (852.30 TRY per vial)

	Hip & knee Arthroplasty	CABG
Surgeries	77,780	12,237
Patients with preoperative anemia	25,667	4038
Patients with IDA	12,834	2019
Treated with FCM	**6417**	**1010**

Postoperative Adverse Event	Cost (TRY)
Sepsis with pneumonia	16,349.93
Sepsis without pneumonia	13,622.70
Acute renal failure	465,226.83
Acute MI	35.975,10
Acute stroke	102.773,81

Table 2. Cost-Effectiveness results of implementing PBM versus no PBM in Turkey

Type of Surgery	Comparators	Cost (TRY)	Incremental cost (TRY)	Avoided adverse events	Incremental avoided adverse events	ICER
Non-cardiac surgery	PBM	13,285	-12,122	1768	1768	PBM dominates
	Control	25,407		0		
Cardiac surgery	PBM	7,417	-11,449	1244	1244	PBM dominates
	Control	18,866		0		

Table 3. Estimation of potential net cost-savings of implementing PBM in Turkey

	Adverse events		Cost of treating adverse events (TRY)		Difference (TRY)
	Control	PBM	Control	PBM	
Sepsis with pneumonia	711	100	11,624,318	1,636,926	9,987,391
Sepsis without pneumonia	529	267	7,201,916	3,637,015	3,564,901
Acute renal failure	258	127	120,025,886	59,108,758	60,917,128
Acute MI	382	289	13,758,103	10,387,831	3,370,272
Acute stroke	101	64	10,426,814	6,598,535	3,828,279
Total	**1982**	**847**	**163,037,036**	**81,369,065**	**81,369,065**
Total cost of PBM					**10,938,163**
Total net cost saving to the SSI (TRY)					**70,729,809**
Total net cost saving to the SSI (€)[c]					**3,844,011**

REFERENCES. 1.Althoff FC et al. Multimodal Patient Blood Management Program Based on a Three-pillar Strategy: A Systematic Review and Meta-analysis. *Ann Surg.* 2019;269(5):794-804. 2.Freedman et al Experience of a network of transfusion coordinators for blood conservation (Ontario Transfusion Coordinators [ONTraC]). *Transfusion.* 2008;48(2):237-250. 3.Liumbruno GM et al Post-operative blood salvage in patient blood management: is it really cost-effective and safe? *Blood Transfus.* 2013;11(2):175-177. 4.Abdullah HR, et al Getting patient blood management Pillar 1 right in the Asia-Pacific: a call for action. *Singapore Med J.* 2020;61(6):287-296. 5.Bernard AC et al Intraoperative transfusion of 1 U to 2 U packed red blood cells is associated with increased 30-day mortality, surgical-site infection, pneumonia, and sepsis in general surgery patients. *J Am Coll Surg.* 2009;208(5):931-937, 937 e931-932; discussion 938-939. 6.Chatterjee S et al Association of blood transfusion with increased mortality in myocardial infarction: a meta-analysis and diversity-adjusted study sequential analysis. *JAMA Intern Med.* 2013;173(2):132-139.7.Jakobsen CJ et al Transfusion of blood during cardiac surgery is associated with higher long-term mortality in low-risk patients. *Eur J Cardiothorac Surg.* 2012;42(1):114-120. 8.Kleineruschkamp A, et al A model-based cost-effectiveness analysis of Patient Blood Management. *Blood Transfus.* 2019;17(1):16-26. 9. Turkish Ministry of Health and European Union. Technical Assistance for Improving the Blood Transfusion management System in Turkey. https://hastakanyonetimisaglikgovtr/. 2019. 10. Unal D et al. Peri-operative blood transfusion in elective major surgery: incidence, indications and outcome - an observational multicentre study. *Blood Transfus.* 2020;18(4):261-279. 11. Drabinski T et al Estimating the Epidemiological and Economic Impact of Implementing Preoperative Anaemia Measures in the German Healthcare System: The Health Economic Footprint of Patient Blood Management. *Adv Ther.* 2020;37(8):3515-3536.

DISCLOSURES. This study was supported by CSL Vifor

Poster presented at ISPOR Europe 2022, 6th-9th November 2022, Vienna, Austria